Cognitive effects of individual enantiomers of LB-102

A. R. Vaino[1], A. Eramo[1], B Grayson[2], V. T. Grattan[1], M. Harte[2,] Z. Prensky[1]

[1]LB Pharmaceuticals Inc., New York, NY, USA, [2] [1]Division of Pharmacy & Optometry, School of Health Sciences, University of Manchester, M13 9PT, UK

Introduction: LB-102 is a novel, racemic, benzamide known to inhibit both dopamine $D_{2/3}$, and 5-HT_7 receptors designed to treat schizophrenia. Modulation of 5-HT_7 has been implicated as affecting cognition and overall well-being and may contribute to improvements in bipolar depression and cognitive impairment associated with schizophrenia. Previous work with LB-102[1] demonstrated that the *R* enantiomer (LB-104) showed a preference for 5-HT_7 binding (K_i v. $D_{2/3}$ = 14.4 nM, K_i v. 5-HT_7 = 15.6 nM) over the *S* enantiomer (LB-103) binding (K_i v. $D_{2/3}$ = 0.4 nM, K_i v. 5-HT_7 = >1000 nM). To establish if the differential binding profiles of the individual enantiomers of LB-102 had effects on behavior in rats, two common tests of cognition/negative symptoms, Novel Object Recognition (NOR) and Social Interaction (SI) were carried out in the sub-chronic phencyclidine model (scPCP) with each enantiomer along with the racemic mixture.

LB-103

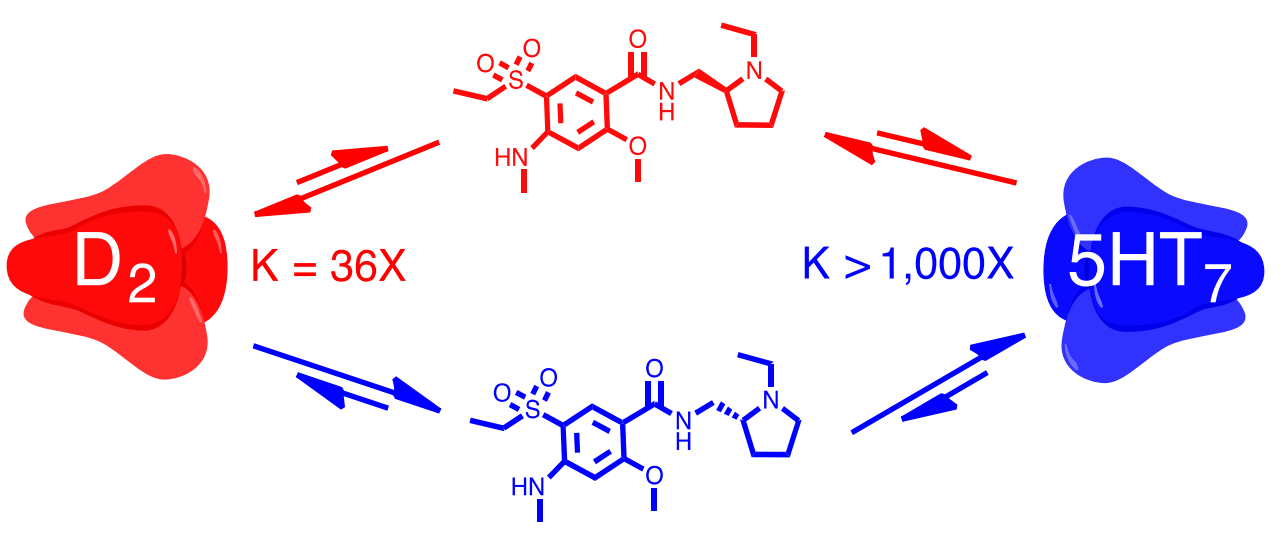
D_2
K = 36X
K > 1,000X
5HT_7

LB-104

- **NOR** Object exploration data from a scPCP NOR study in rats (2 mg/kg i.p. twice daily followed by 7 days drug free (n = 12/grp) measured 60-180 min post drug. Rats were dosed with 5, 15, or 45 mg/kg LB-102, LB-103, or LB-104 PO.
- **SI** Sniffing index data from a sub-chronic PCP SI study in rats (2 mg/kg i.p. twice daily followed by 7 days drug free (n = 12/grp) measured 60-180 min post drug. Rats were dosed with 5, 15, or 45 mg/kg LB-102, LB-103, or LB-104 PO.
- **Plasma PK in rats:** PK samples were taken from rats (n = 3/grp) at 0.0416, 0.083, 0.166, 0.333, 0.5, 1, 2, 4, 8 and 16 hours after oral dosing and analyzed by HPLC to determine if T_{max} for either enantiomer was different from the other.

Results:

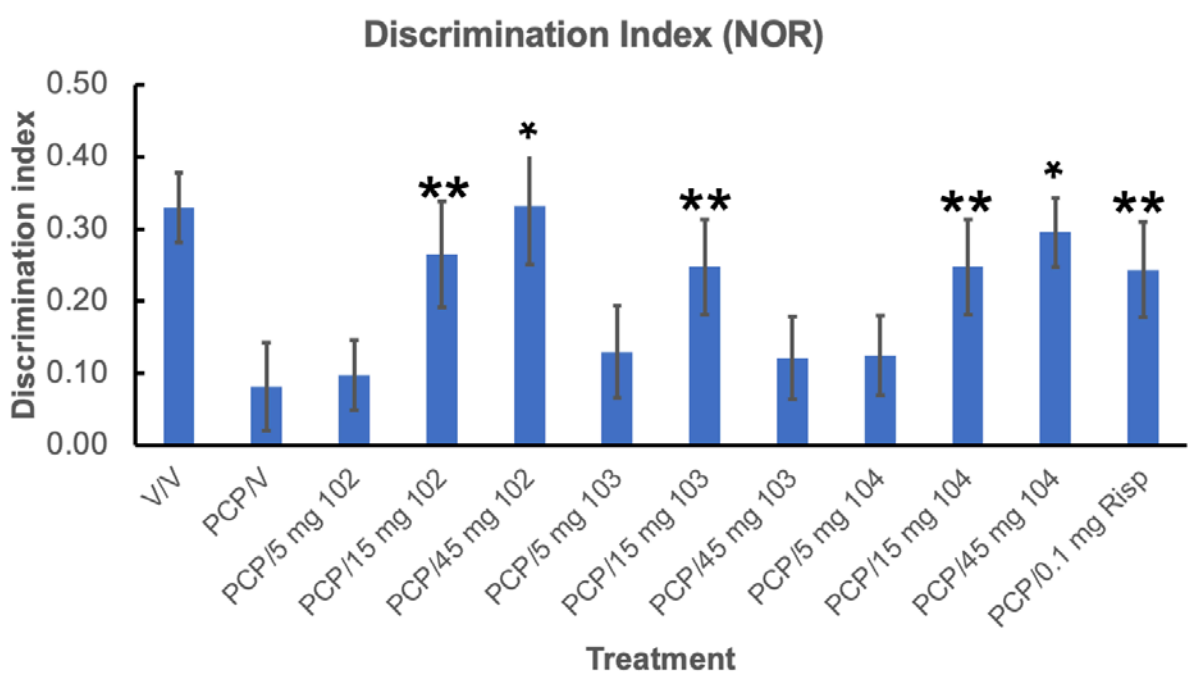
Discrimination Index (NOR)
Discrimination index
0.50
0.40
0.30
0.20
0.10
0.00
V/V
PCP/V
PCP/5 mg 102
PCP/15 mg 102
PCP/45 mg 102
PCP/5 mg 103
PCP/15 mg 103
PCP/45 mg 103
PCP/5 mg 104
PCP/15 mg 104
PCP/45 mg 104
PCP/0.1 mg Risp
Treatment

* p < 0.05 v PCP ** p < 0.1 v PCP

- 45 mg/kg LB-102 and LB-104 had p < 0.05 v 45 mg/kg LB-103

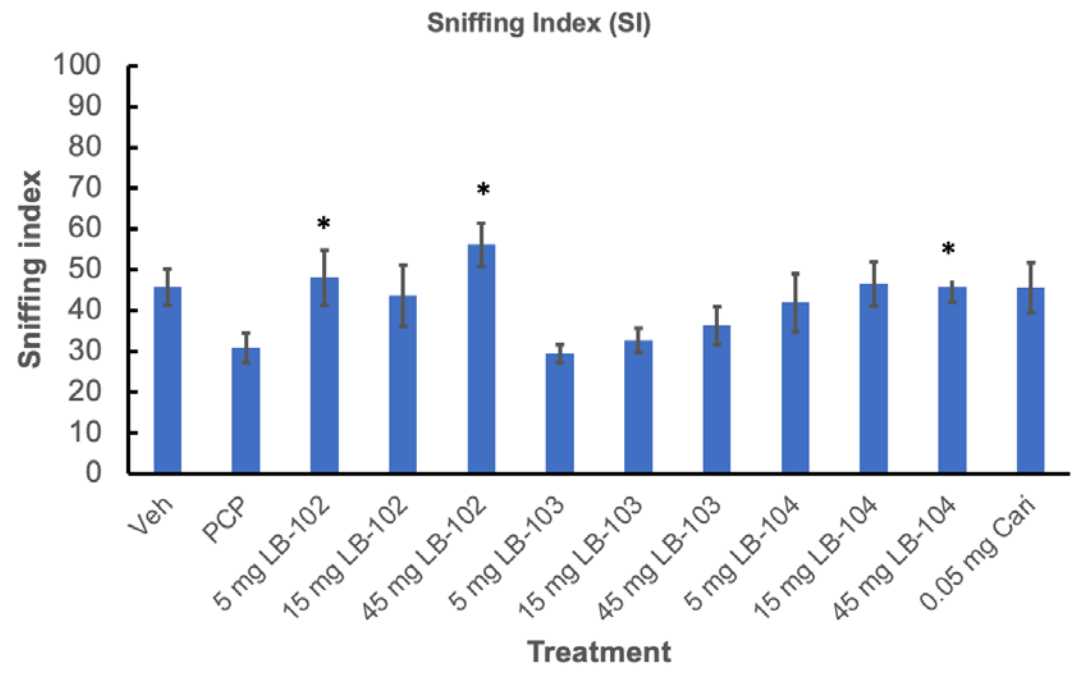
Sniffing Index (SI)
Sniffing index
100
90
80
70
60
50
40
30
20
10
0
Veh
PCP
5 mg LB-102
15 mg LB-102
45 mg LB-102
5 mg LB-103
15 mg LB-103
45 mg LB-103
5 mg LB-104
15 mg LB-104
45 mg LB-104
0.05 mg Cari
Treatment

* p < 0.05 v PCP

- LB-102 = LB-104 at all doses (p > 0.1), LB-104 > LB-103 (p < 0.05) at 15 and 45 mg/kg
- LB-102 > LB-103 at all doses (p < 0.05) at 5 and 45 mg/kg (p < 0.1) at 15 mg/kg

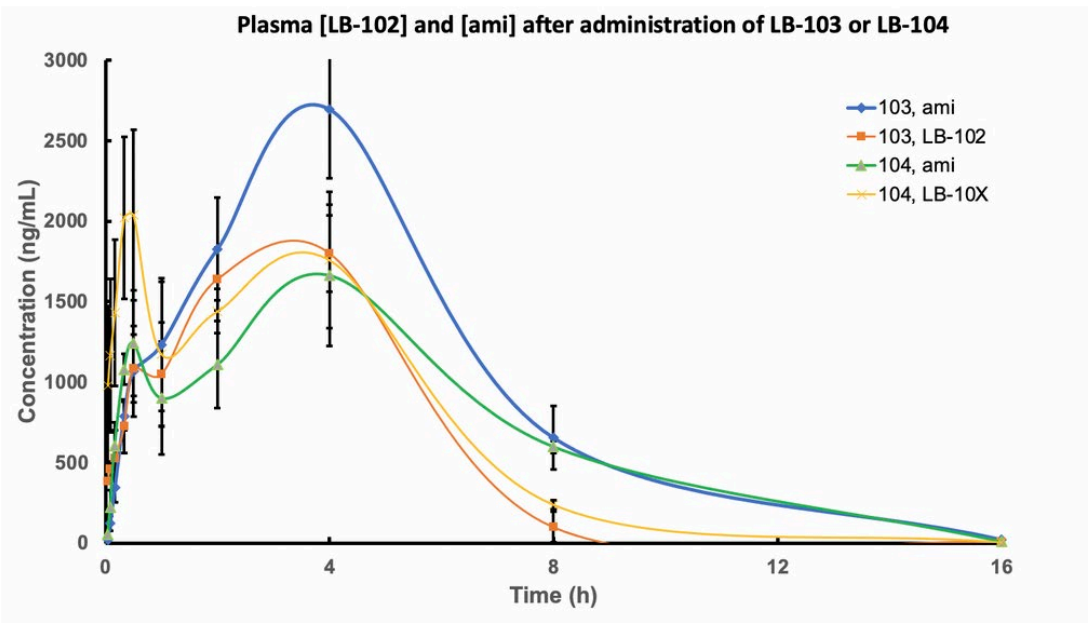
Plasma [LB-102] and [ami] after administration of LB-103 or LB-104
Concentration (ng/mL)
3000
2500
2000
1500
1000
500
0
0
4
8
12
16
Time (h)
103, ami
103, LB-102
104, ami
104, LB-10X

- Plasma PK profile LB-102 and amisulpride in rats (n = 3) after administration of a single 100 mg/kg oral dose of either LB-103 or LB-104

- ◆ In two models of cognition LB-102 (racemic) was equivalent to LB-104 (*R* enantiomer) in the scPCP model
- ◆ LB-102 and LB-104 were superior to LB-103 (*S* enantiomer) at certain doses
- ◆ These results suggest that the increased affinity of LB-104 for 5-HT_7 manifests in behavioral assays but that the magnitude is subtle enough that 2X greater K_i is not noticeable
- ◆ A relative decrease in dopamine binding with LB-104 may improve tolerability, though interrogating safety in animal studies can be challenging

Disclosures

This work was funded by LB Pharmaceuticals. AE, ZP, and AV are employees of LB Pharmaceuticals. VG is a consultant to LB Pharmaceuticals. AE, VG, ZP, and AV are shareholders of LB Pharmaceuticals.

1 V. Grattan, A. R. Vaino, Z. Prensky, and M. S. Hixon, "Antipsychotic benzamides amisulpride and LB-102 display polypharmacy as racemates, S enantiomers engage receptors D_2 and D_3, while R enantiomers engage 5-HT_7", ACS *Omega,* **2019**, *4*, 14151–14154