Safety, pharmacokinetics, and pharmacodynamics of LB-102, a selective D2 / 5-HT7 antagonist, in healthy volunteers

Andrew R. Vaino,[1] Vincent T. Grattan,[1] Zachary Prensky,[1] Mark S. Hixon,[2] Lukasz Biernat[3]

[1]LB Pharmaceuticals Inc., New York, NY, [2]Mark S. Hixon Consulting LLC, San Diego, CA, [3]Medpace Inc., Cincinnati OH

33rd ECNP Congress Virtual 12-15 September 2020

Introduction and Background

Despite 23 FDA approved drugs to treat schizophrenia the majority of patients with the disorder are not adequately treated. A 2009 report [1] noted that nearly 1/3 of schizophrenia patients switched medication during the course of a year (on average after 100 days). A recent meta analysis encompassing 54,000 patients and 32 schizophrenia drugs [2] demonstrated that amisulpride, a drug unavailable in the United States, was among the most effective and best tolerated schizophrenia drugs.

LB-102 is a, patented, *N*-methylated version of the highly effective antipsychotic amisulpride which has been used for decades in Europe but is unavailable to American schizophrenia patients. LB-102 was designed to improve on the poor blood-brain-barrier permeability of amisulpride.

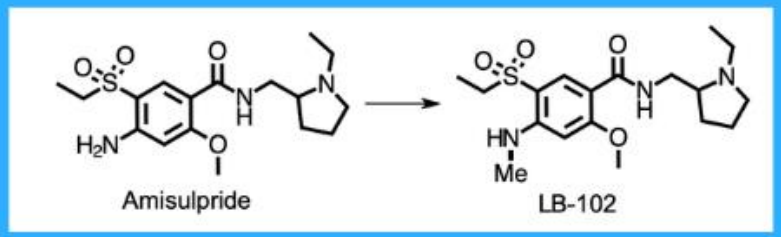
Amisulpride
LB-102

Pre-clinical studies of LB-102, including in vitro CNS receptor binding, PK studies, and animal behavioral studies [3] demonstrated that LB-102 was equivalent, or superior, to amisulpride.

A Phase 1 clinical study (n = 64) in healthy volunteers (NCT04187560) was recently completed at Medpace in Cincinnati. This was a Single Ascending Dose (SAD) /Multiple Ascending Dose (MAD) study in which subjects were randomized 3:1 active:placebo. LB-102 was dosed orally. The primary endpoint of this study was safety, with pharmacokinetics as a secondary endpoint.

Phase 1 Results

Adverse events, Phase 1 study of LB-102

SAD

Dose (mg)	Adverse Event	n	Severity	Related to drug
10	Prolactin elevation	1	Mild	Definitely
	Abdominal cramps	1	Mild	Unrelated
50	Prolactin elevation	5	Mild	Definitely
	Diarrhea	1	Mild	Unlikely
	Upper respiratory infection	1	Mild	Unrelated
100	Prolactin elevation	1	Mild	Definitely
	Urticaria	1	Mild	Possibly
	Headache	1	Mild	Probably
	Upper respiratory infection	1	Mild	Unrelated
	Nausea	1	Mild	Probably
150	Prolactin elevation	1	Mild	Definitely
	Low back pain	1	Mild	unrelated
200	QT prolongation	1	Mild	Definitely
	Acute dystonia	1	Moderate	Definitely
	Palpitations	1	Mild	Unlikely
	Nausea	1	Mild	Possibly
	Gastroesophageal reflux	1	Mild	Possibly
	Insomnia	1	Mild	Probably
	Sore throat	1	Mild	Unrelated
	Headache	1	Mild	Unrelated

MAD

Dose (mg, BID)	Adverse Event	n	Severity	Related to drug
50	Dizziness	1	Mild	Unrelated
	Prolactin elevation	2	Mild	Definitely
75	Elevated prolactin	2	Mild	Definitely
	Abdominal cramps	1	Mild	Unrelated
	Migraine headache	1	Moderate	Possibly
	Acute dystonic reaction	1	Moderate	Definitely
	Intermittent dizziness	1	Mild	Probably
	Intermittent nausea	1	Mild	Probably
	Intermittent drowsiness	1	Mild	Probably
	Bug bite	1	Mild	Unrelated
	Insomnia	1	Mild	Possibly
	Low back pain	1	Mild	Unrelated
100	Acute dystonia	2	Moderate	Definitely
	Drowsiness	1	Mild	Unrelated
	Dry mouth	1	Mild	Unrelated
	Elevated prolactin	1	Mild	Definitely
	Nausea	1	Moderate	Probably
	Emesis	1	Mild	Possibly

LB-102 was well-tolerated at up to 75 mg BID over a week.

Two instances of EPS (acute dystonia), consistent with dopamine receptor occupancy > 80% [5], were noted in the 100 mg BID cohort and one instance was reported in the 75 mg BID cohort.

Superimposing LB-102 plasma C_{max} on published [6] plasma concentrations of amisulpride versus dopamine percent receptor occupancy:

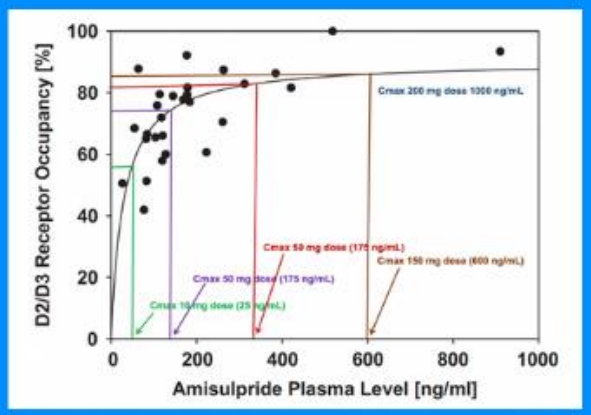
D2/D3 Receptor Occupancy [%]
Amisulpride Plasma Level [ng/ml]
Cmax 200 mg dose 1000 ng/mL
Cmax 50 mg dose (175 ng/mL)
Cmax 150 mg dose (600 ng/mL)
Cmax 50 mg dose (175 ng/mL)
Cmax 10 mg dose (25 ng/mL)

suggests that doses of LB-102 as low as 50 to 100 mg/day could effectively treat schizophrenia.

Pharmacokinetics

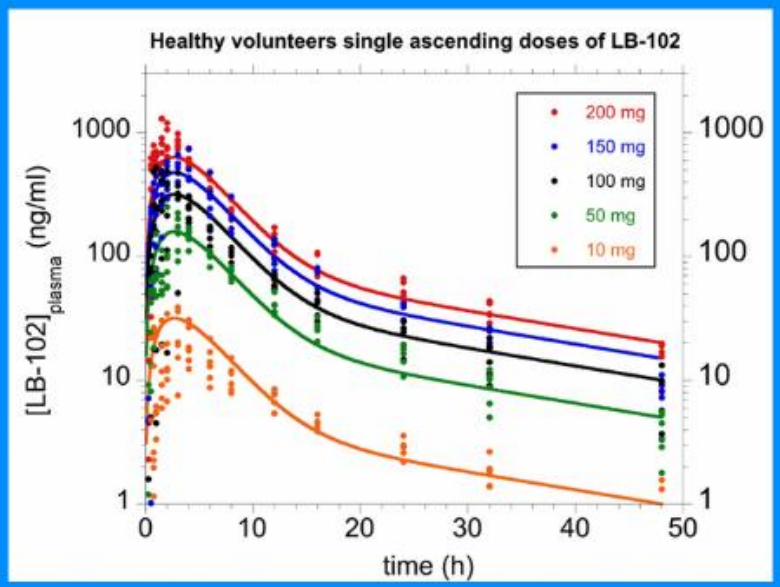
Healthy volunteers single ascending doses of LB-102
200 mg
150 mg
100 mg
50 mg
10 mg
[LB-102]plasma (ng/ml)
time (h)

In the SAD PK study, overall plasma concentration of LB-102 was ~2.5X greater than expected based on comparison to published amisulpride data [4].

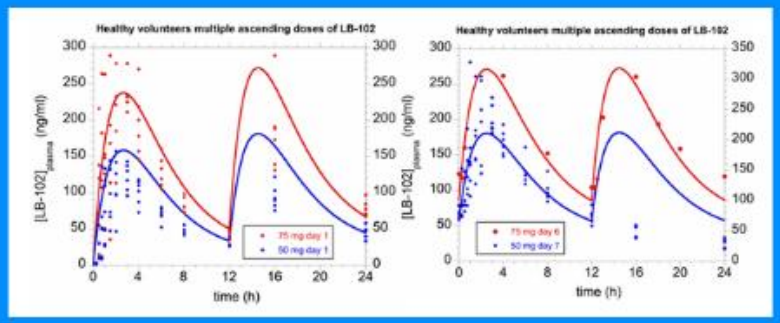
Healthy volunteers multiple ascending doses of LB-102
75 mg day 1
50 mg day 1
Healthy volunteers multiple ascending doses of LB-102
75 mg day 6
50 mg day 7
[LB-102]plasma (ng/ml)
time (h)

Conclusions and Next Steps

- LB-102, a novel methylated version of amisulpride (an effective schizophrenia drug used in Europe for decades), was well-tolerated at up to 75 mg BID in a one week study
- LB-102 displayed dose-linear PK up to 150 mg as a single dose, and has a PK profile suitable for oral administration
- Using EPS as a biomarker suggests that dosing LB-102 at 75 mg BID approaches 80% dopamine receptor occupancy, just above the 60% to 75% typically observed in schizophrenia drugs [6]
- Based on the above, 50 to 100 mg/day LB-102 could provide an effective treament for schizophrenia (amisulpride is typically dosed 400 to 800 mg/day)
- A PET study to measure dopamine receotor occupancy directly in humans is being planned, and will inform dosing in a Phase 2 clinical study in schizophrenia patients planned for 2021